Real-world Outcomes Associated with Lomitapide Use in Patients with Homozygous Familial Hypercholesterolemia: A Retrospective Database Analysis

Ruo-Ding Tan[1], Qinghao Peng[1], Christian Frois[1], Taylor Salinardi[2], Eliot A. Brinton[3]

[1]Analysis Group, Boston, MA, USA; [2]Aegerion Pharmaceuticals, Boston, MA, USA; [3]Utah Lipid Center, Salt Lake City, UT, USA

Background

- Homozygous familial hypercholesterolemia (HoFH) is a heterogeneous ultra-rare genetic disorder, affecting an estimated 1:1 000 000 to 1:160 000 individuals.[1]
- Patients typically manifest with extremely elevated levels of low-density lipoprotein cholesterol (LDL-C) leading to development of premature major adverse cardiovascular events (MACE). If left untreated, the expected lifespan for patients with HoFH is less than 30 years.[1] Traditional treatment options often fail to provide adequate LDL-C lowering and/or LDL-C goal achievement.
- Lomitapide, a microsomal triglyceride transfer protein inhibitor, was approved for LDL-C lowering in HoFH in the United States in 2012 based on data from 29 patients enrolled in a single-arm, open-label, phase 3 trial.[2] Data from the phase 3 trial showed that patients receiving once-daily oral lomitapide achieved a mean 50% lowering of LDL-C levels from baseline at week 26, the study's primary endpoint.
- Despite lomitapide being indicated as an adjunct therapy to diet and other lipid-lowering treatments for HoFH, no randomized trials, few real-world studies, and no published lomitapide studies address MACE in HoFH, which reflects the ultra-rare prevalence of HoFH. This study presents for the first time MACE rates in lomitapide-treated patients.

Objectives

- Integrate real-world data for lomitapide collected from manufacturer's patient support program and an administrative claims database to create an enriched dataset
- Assess LDL-C, annual MACE and hospitalization rates before initiation, during lomitapide treatment, and 1 year after lomitapide discontinuation in patients who stopped taking it

Methods

Data Sources

- Optum Clinformatics Data Mart (CDM)
 - De-identified claims data (January 2012 through March 2016) for over 25 million covered lives annually from commercial and Medicare health plans
 - LDL-C (LOINC code 02089-1) was available for claims from a limited number of vendors
- Manufacturer's (Aegerion) COMPASS database
 - Anonymous data collected by the manufacturer as part of its patient support program for all patients treated by lomitapide
 - Patient-level information, including age, gender, health plan, other medications, lomitapide prescription(s) shipment date(s) and state, and LDL-C levels
- Patient-level matching and pooling between CDM and COMPASS (**Figure 1**)
 - De-identified data were matched based on
 - Patient characteristics (year of birth/age, gender, state), and
 - First and last drug fill and shipment dates (CDM and COMPASS, respectively)
 - LDL-C test results from COMPASS were added to the analysis to complement LDL-C data from CDM

Figure 1: *Patient-Level Matching Between CDM and COMPASS*

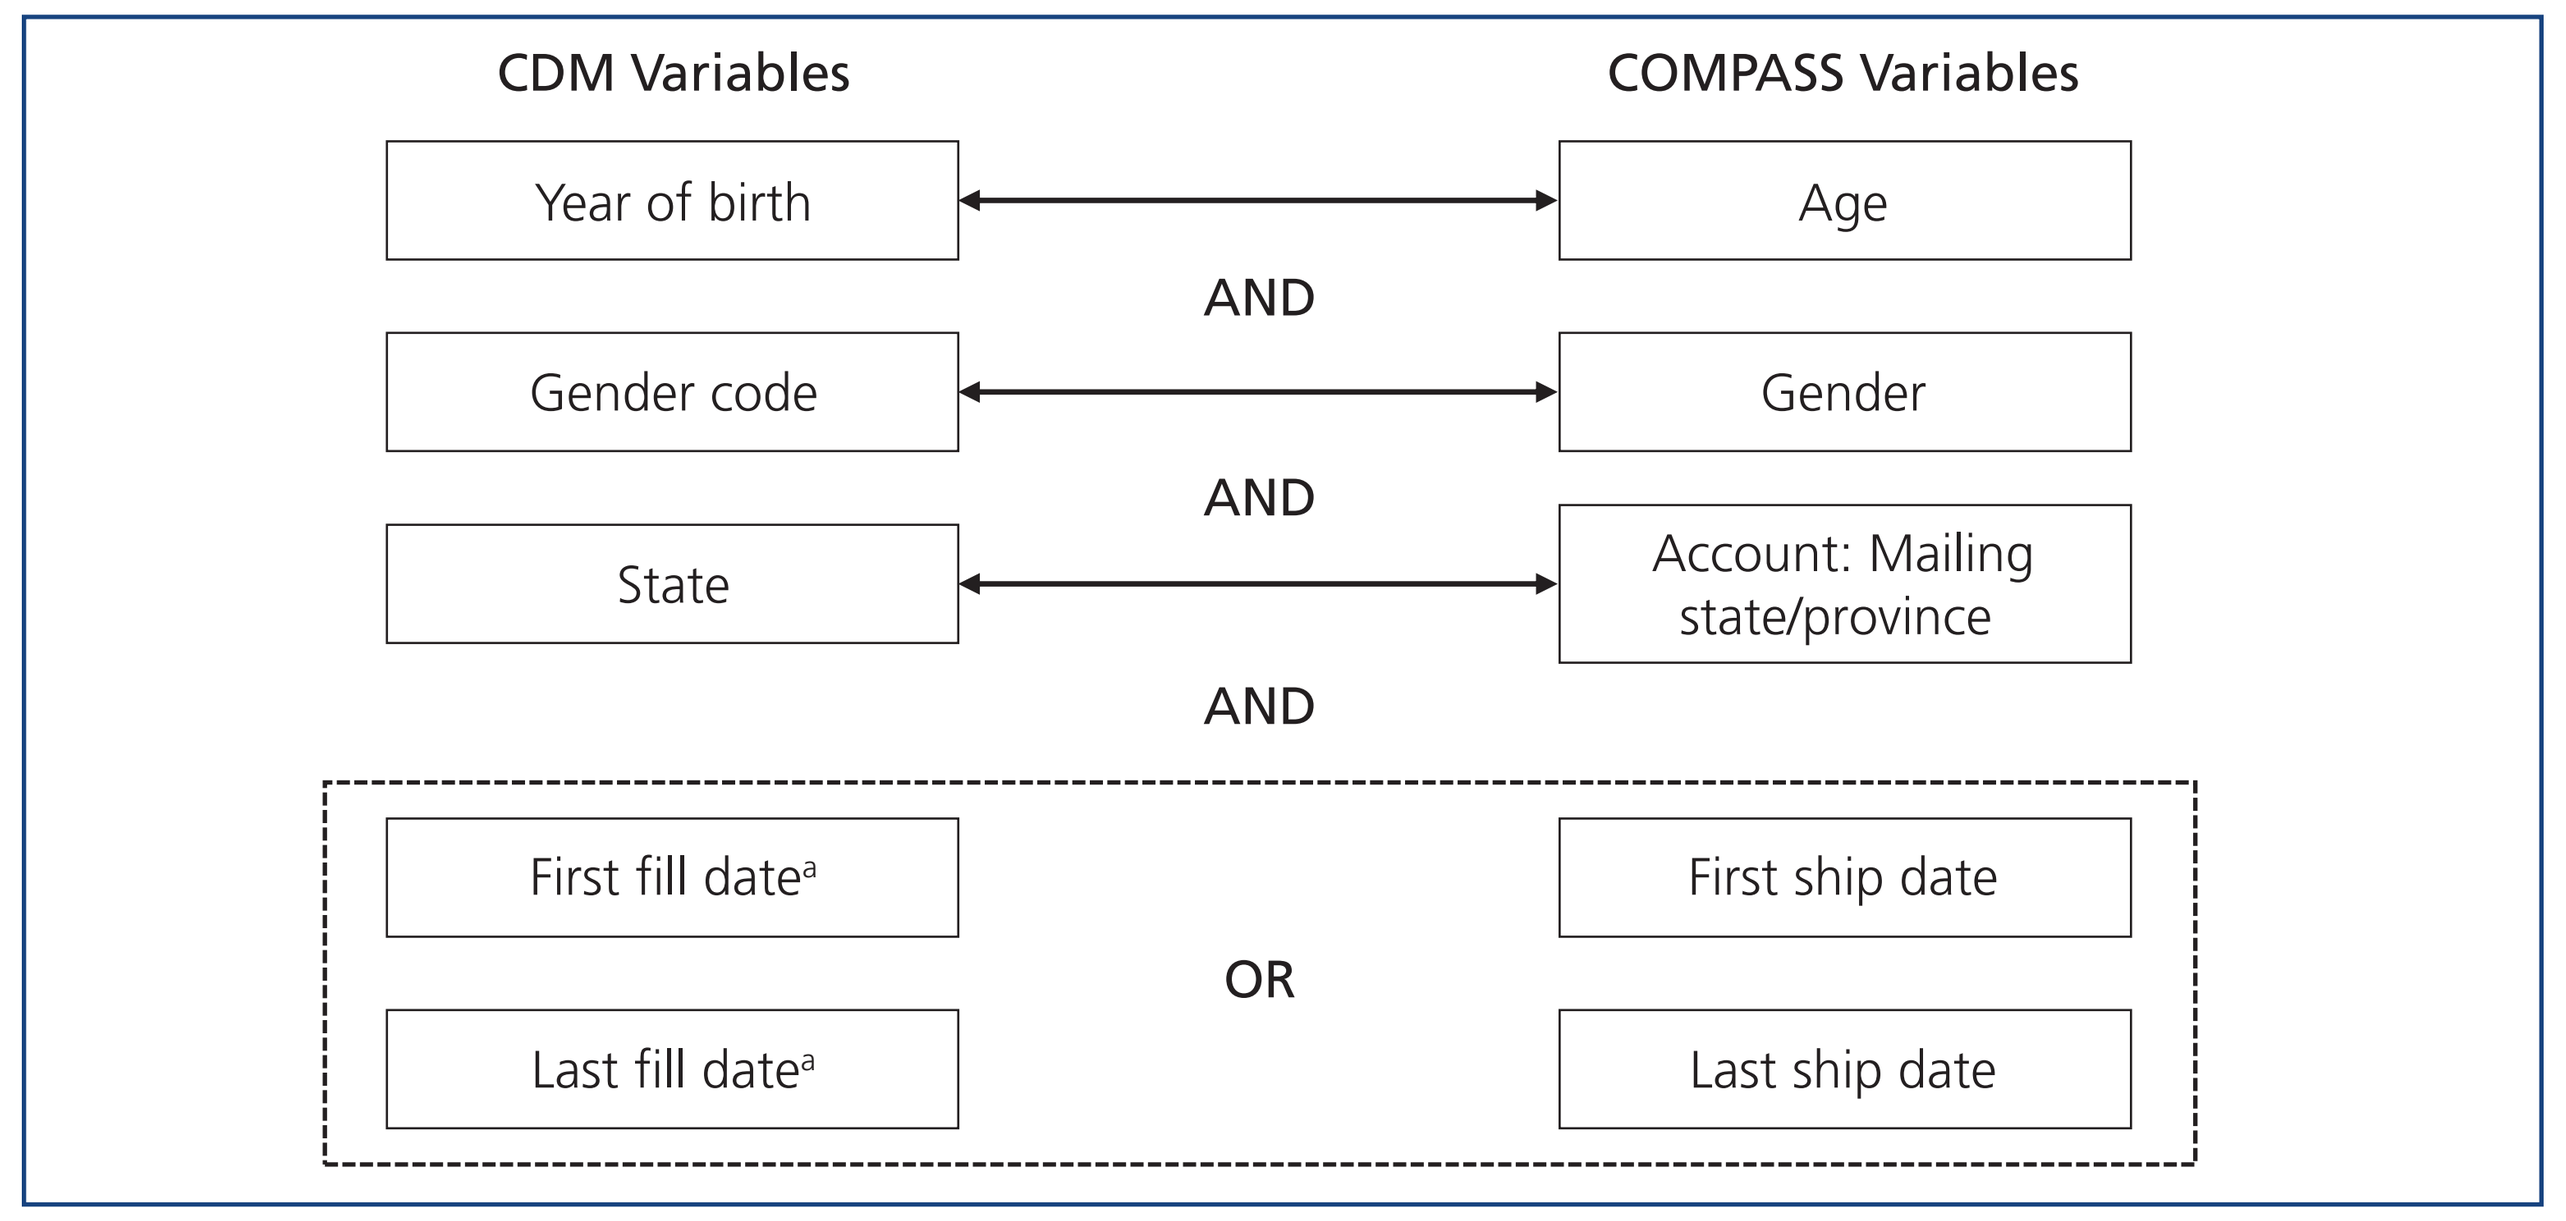
CDM Variables
COMPASS Variables
Year of birth
Age
AND
Gender code
Gender
AND
State
Account: Mailing state/province
AND
First fill date[a]
First ship date
OR
Last fill date[a]
Last ship date

Abbreviation: CDM, Optum Clinformatics Data Mart.
[a] First and last fill dates were calculated based on all lomitapide prescription drug claims as observed for each individual patient.

Subject Inclusion Criteria

- At least 180 days of continuous enrollment in medical and pharmacy plans before the index date (date of first lomitapide prescription)
- ≥ 18 years of age on the index date
- Absence of cancer (except non-melanoma skin cancer) during the study follow-up period (index date to the end of the data cut or end of enrollment eligibility)
- At least 1 pre-index and 1 follow-up LDL-C value

Study Measures

- Study outcomes included change in LDL-C levels, MACE and hospitalization rates
 - Change in LDL-C levels was measured before lomitapide initiation, during lomitapide treatment, and after lomitapide discontinuation
 - MACE occurrence, defined as acute myocardial infraction, acute ischemic stroke, or coronary revascularization, was analyzed based on diagnosis codes (ICD-9-CM, ICD-10-CM) or procedure codes (HCPCS and CPT)
 - Hospitalization was identified based on claims records associated with an inpatient stay in CDM data
- Patients metrics:
 - LDL-C responsive to lomitapide (sustained responder)
 - Mean LDL-C reduction from pre-index level of at least 10%, and
 - Post-index LDL-C observed over a period greater than 45 days from the pre-index reading, and
 - Individual post-index readings no higher than 30% of pre-index level
 - Discontinuation: at least 60 days between the last lomitapide fill being consumed and the data cut point or end of enrollment eligibility

Descriptive and Statistical Analysis

- All clinical outcomes were compared before and after lomitapide initiation, as well as before and after lomitapide discontinuation. Statistical significance of these comparisons was assessed using paired t-tests.

Results

Patient Selection

- 35 patients had pre-index and follow-up LDL-C values in the CDM database
- 27 additional patients had additional LDL-C results from COMPASS providing study eligibility
- Final analysis set was comprised of both groups, totaling 62 patients (**Figure 2**)

Figure 2: *Patient Selection*

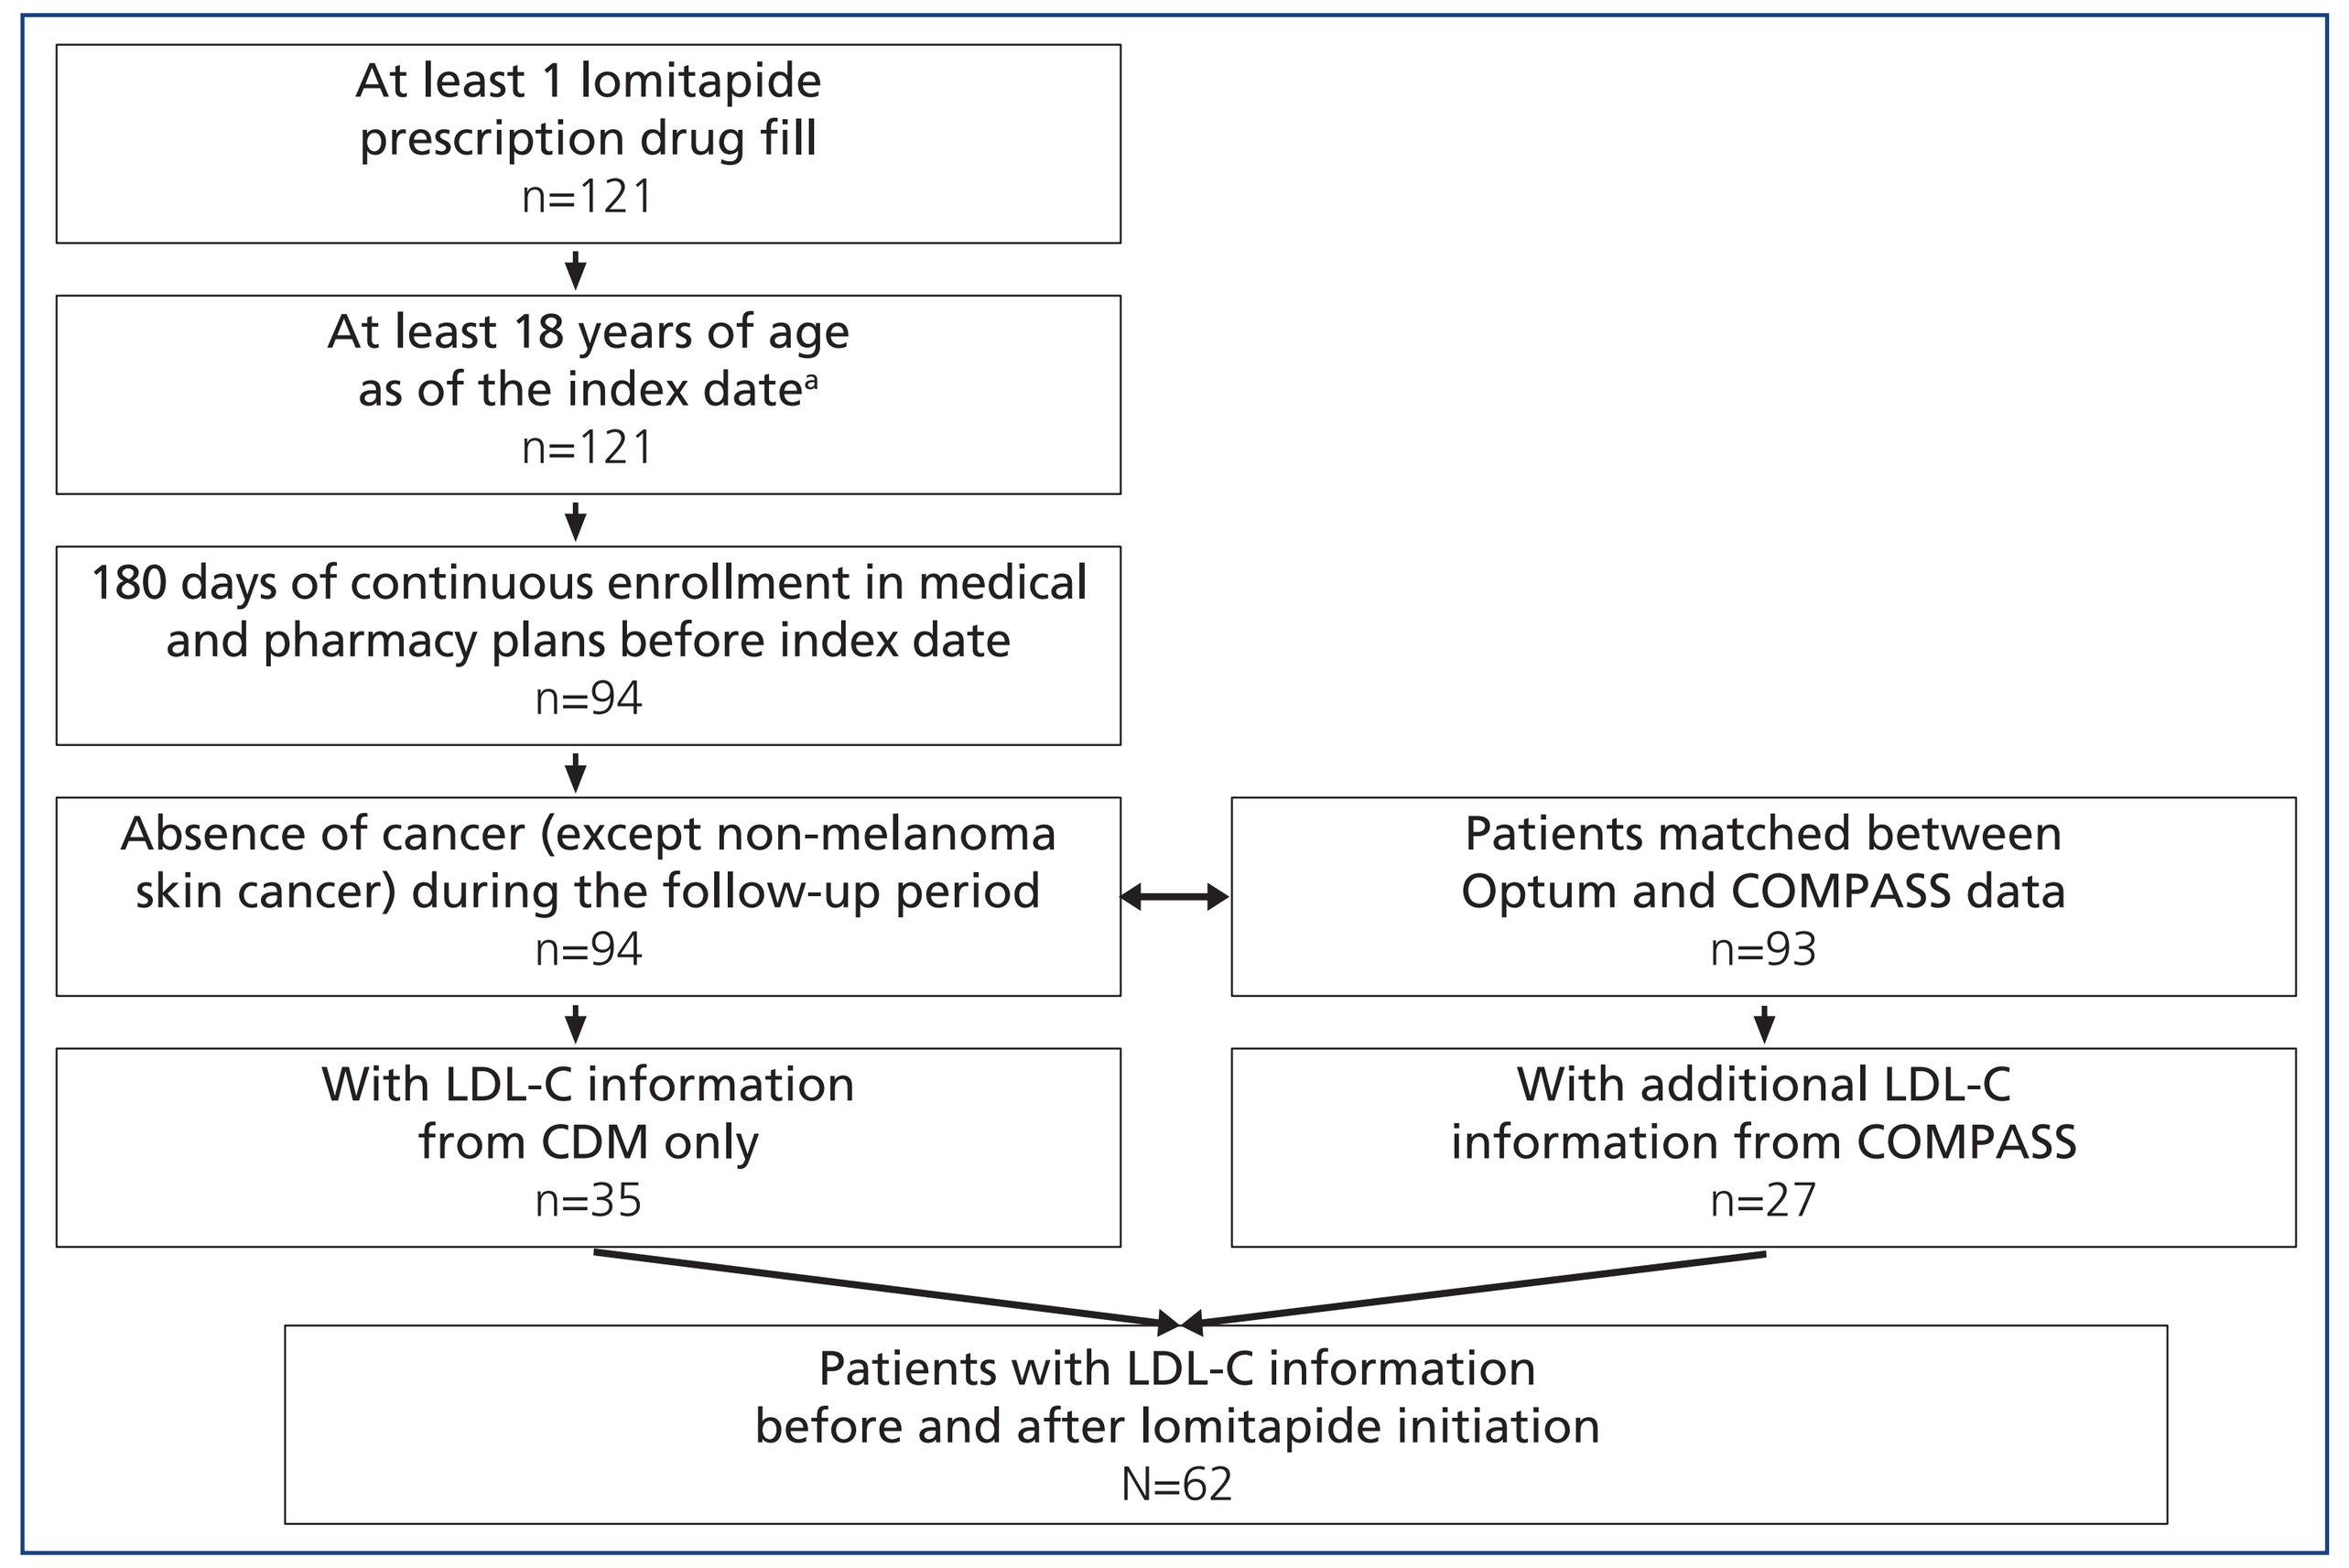
At least 1 lomitapide prescription drug fill
n=121
At least 18 years of age as of the index date[a]
n=121
180 days of continuous enrollment in medical and pharmacy plans before index date
n=94
Absence of cancer (except non-melanoma skin cancer) during the follow-up period
n=94
Patients matched between Optum and COMPASS data
n=93
With LDL-C information from CDM only
n=35
With additional LDL-C information from COMPASS
n=27
Patients with LDL-C information before and after lomitapide initiation
N=62

Abbreviations: LDL-C, low-density lipoprotein-cholesterol
[a] Index date is the date of the first lomitapide prescription drug fill.

Patient Characteristics

- The mean age for the final analysis set was 58 years (SD, 12) at the index date, and 36 of the 62 patients were female
- Most patients (n=50/62, 81%) used statins before initiating lomitapide, and nearly half (n=27/62, 44%) used ezetimibe. No patients used PCSK9 inhibitors or mipomersen prior to lomitapide initiation

Clinical Outcomes Before and After Lomitapide Initiation

- Lomitapide use was associated with significant LDL-C reductions during the year following lomitapide initiation (–27% at 12 months, *P*<0.001) (**Table 1**)
- There was a trend towards a possibly somewhat lower MACE (–19%, *P*=0.29) and a large decrease in hospitalization (–63%, *P*=0.09) during the year after lomitapide initiation (**Table 1**)
- A majority, 65% (40/62) of patients, had a 10% or greater decrease to LDL-C observed over a period greater than 45 days following the pre-index reading. The average duration of response was 17 months. By definition, in these patients the LDL-C decrease was greater than in the overall study population, 38% (*P*<0.001) vs 27%, respectively. Also, as would be expected in those selected for a greater LDL-C response, both the average number of MACE and hospitalizations per patient per year trended downward more than in the overall HoFH population, by 37% (*P*=0.16) and 75% (*P*=0.06) , respectively (**Table 1** and **Figure 3**)

Table 1: *Clinical Outcomes Before and After Lomitapide Initiation*

	1 Year Before Lomitapide Initiation[b]	1 Year After Lomitapide Initiation[b]	Absolute Change	(95% CI)	Relative Change (%)	P-value
All Patients (N=62)						
Mean LDL-C, mg/dL[c]	199	145	–54	(–68.8, –39.2)	–27%	<.001
Mean number of MACE per patient per year	0.58	0.47	–0.11	(–0.50, 0.28)	–19%	.29
Mean number of hospitalizations per patient per year	0.20	0.07	–0.13	(–0.31, 0.06)	–63%	.09
LDL-C Responsive to Lomitapide[a] Among All Patients (n=40/62)						
Mean LDL-C, mg/dL[c]	208	130	–78	(–92.8, –63.9)	–38%	<.001
Mean number of MACE per patient per year	0.59	0.38	–0.22	(-0.65, 0.21)	–37%	.16
Mean number of hospitalizations per patient per year	0.26	0.06	–0.20	(-0.45, 0.06)	–75%	.06

Abbreviations: LDL-C, low-density lipoprotein-cholesterol; MACE, major adverse cardiac event.
[a] Response to lomitapide was defined as a mean post-index LDL-C level at least 10% lower than the baseline, and post-index LDL-C observed over a period greater than 45 days from the pre-index reading without lomitapide discontinuation. Additionally, individual post-index readings no higher than 30% of pre-index level.
[b] For patients with less a year of observation period before or after lomitapide initiation, study outcomes were annualized to estimate the annual average.
[c] The mean was calculated for patients with LDL-C readings within 1 year before and 1 year after lomitapide initiation. 50 and 33 patients are included in the calculation for all patients and patients with LDL-C responsive to lomitapide, respectively.

Figure 3: *Clinical Outcomes in LDL-C Responders to Lomitapide Treatment (n=40)*

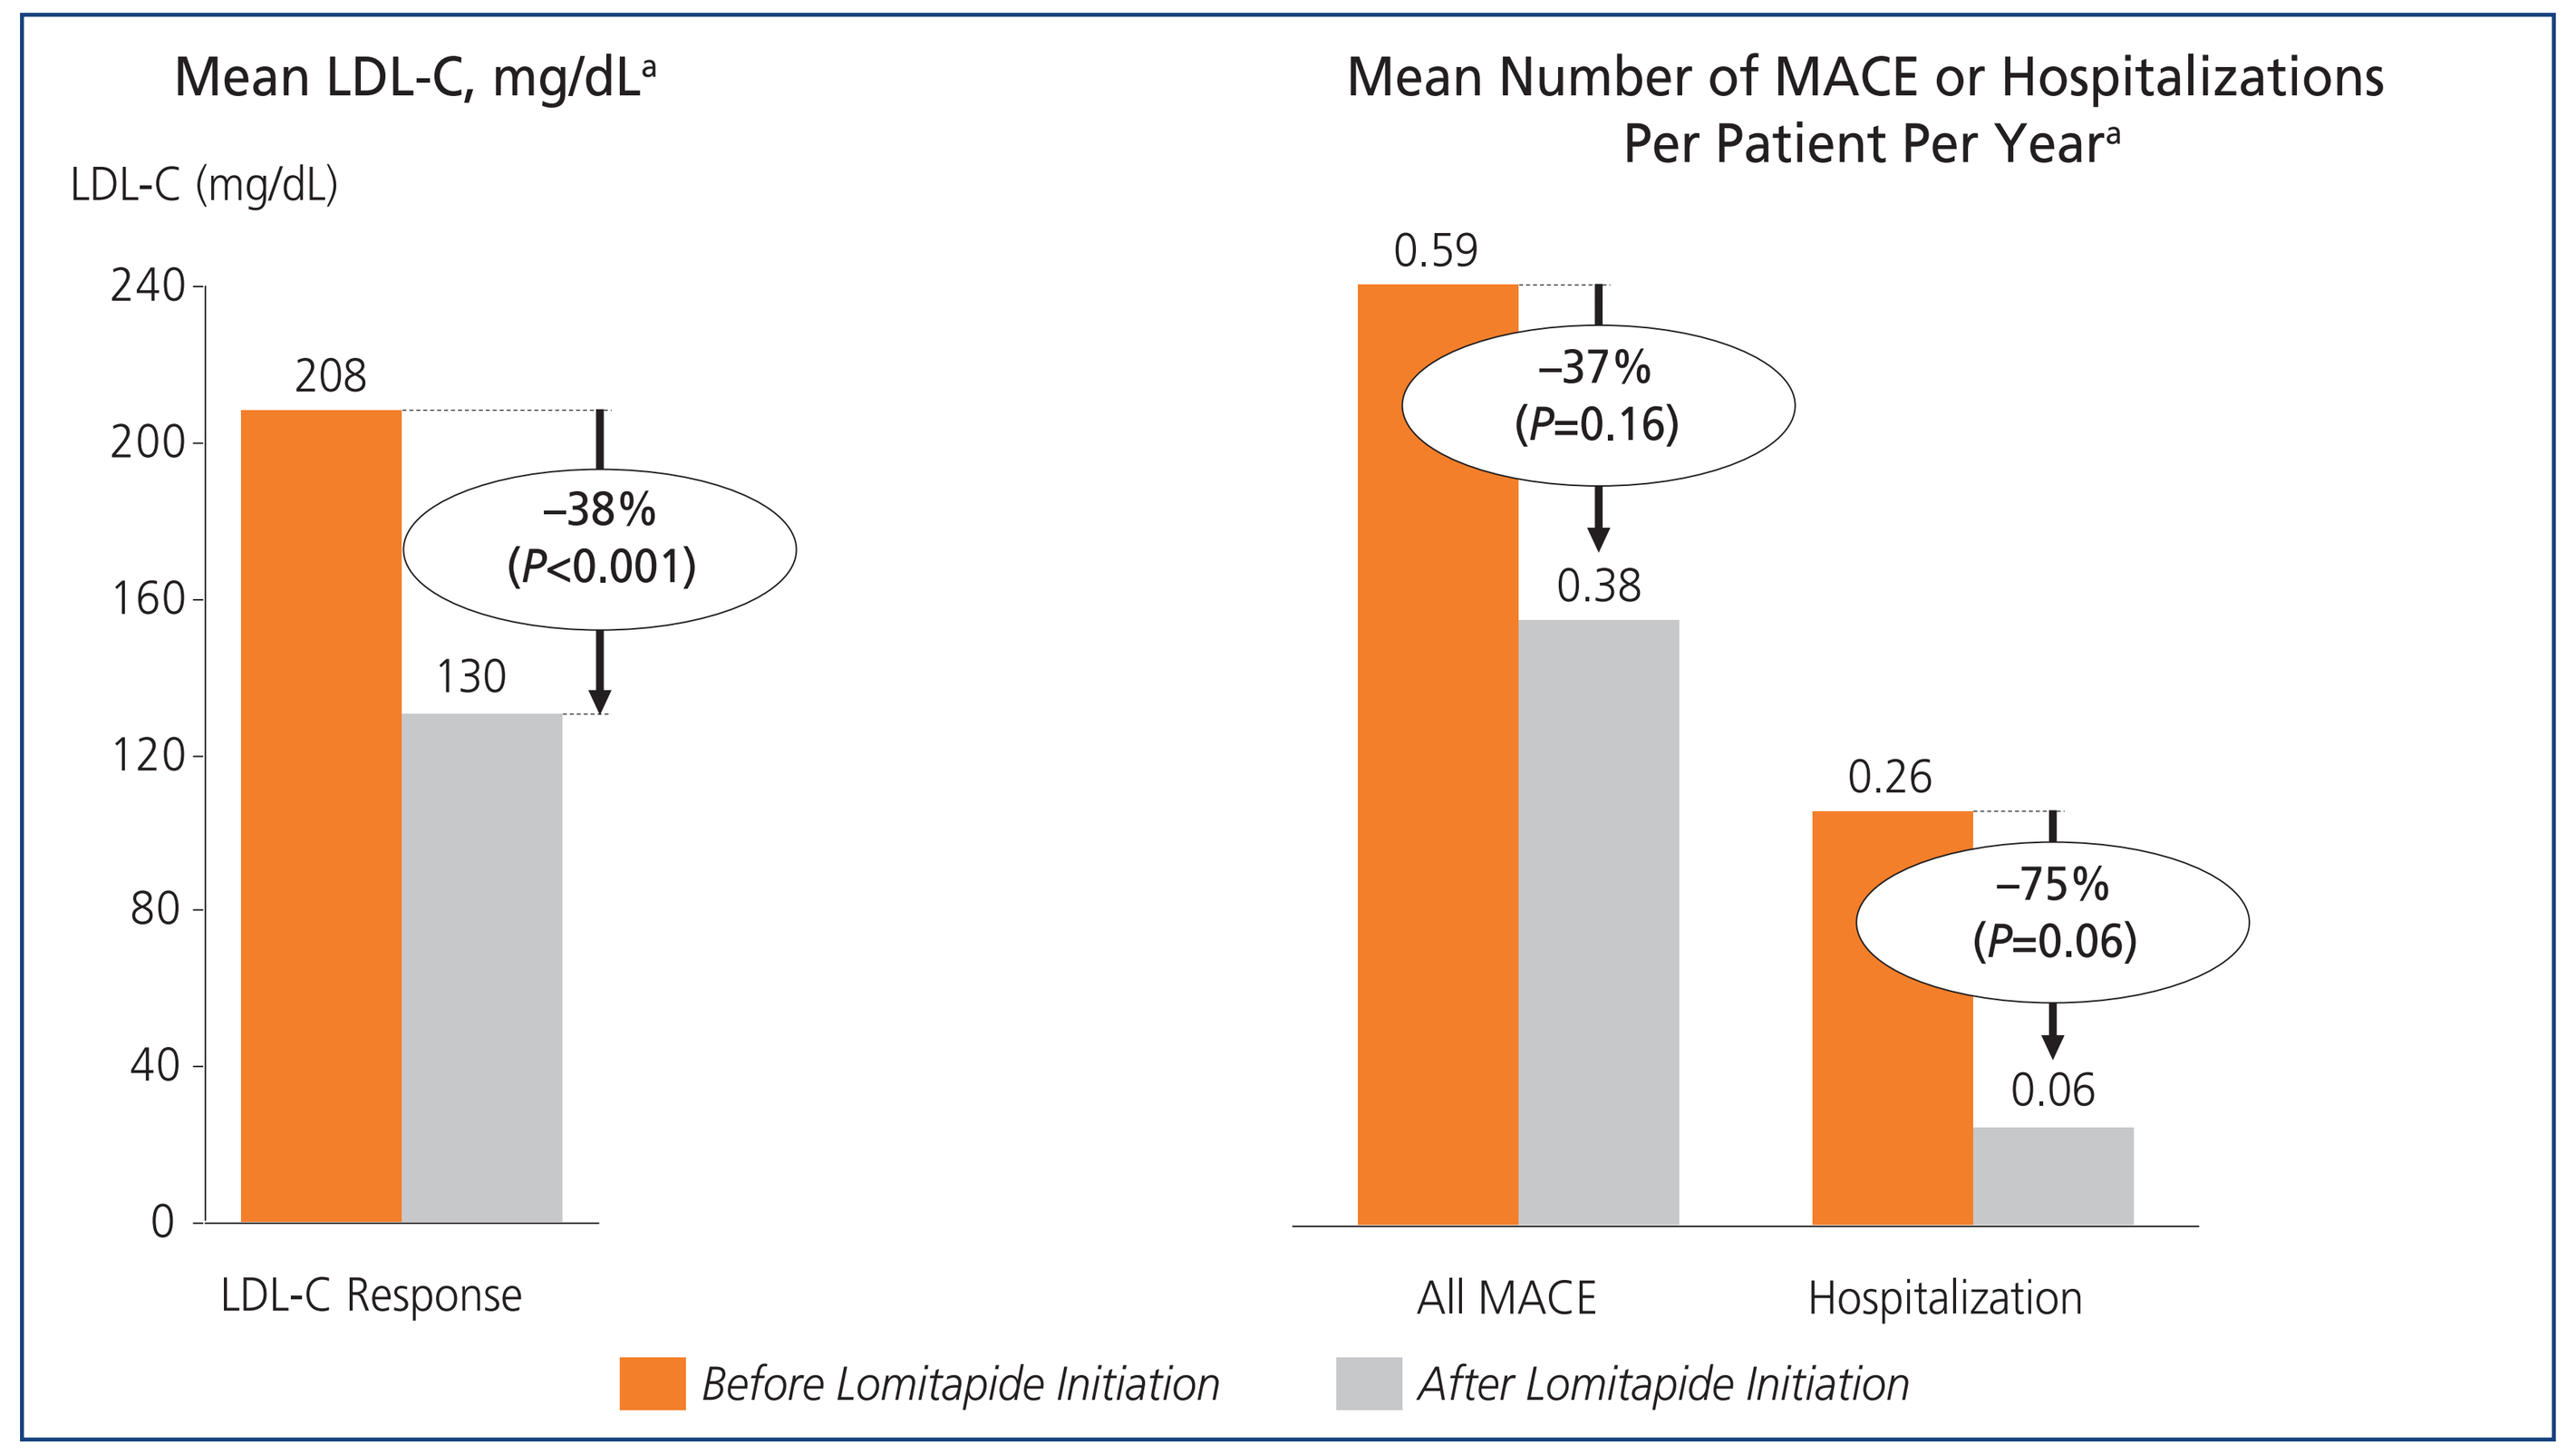
Mean LDL-C, mg/dL[a]
LDL-C (mg/dL)
240
200
160
120
80
40
0
208
130
–38%
(P<0.001)
LDL-C Response
Mean Number of MACE or Hospitalizations Per Patient Per Year[a]
0.59
–37%
(P=0.16)
0.38
0.26
–75%
(P=0.06)
0.06
All MACE
Hospitalization
Before Lomitapide Initiation
After Lomitapide Initiation

Note:
[a] All results compared in year before and after lomitapide initiation.

Clinical Outcomes Before and After Lomitapide Discontinuation

- Among responders who later discontinued lomitapide, outcomes tended to worsen afterwards, with a 20% LDL-C increase (*P*=0.06); 3.5-fold higher annual MACE rate (*P*=0.08); and a 1.2-fold higher annual hospitalization rate (*P*=0.10) (**Table 2** and **Figure 4**)

Table 2: *Clinical Outcomes Before and After Lomitapide Discontinuation*

	1 Year Before Lomitapide Discontinuation[a]	1 Year After Lomitapide Discontinuation[a]	Absolute Change	95% CI	Relative Change (%)	P-value
Discontinuation Following LDL-C Response to Lomitapide (n=27/40)						
Mean LDL-C, mg/dL[b]	120	144	24	(–7.3, 55.5)	20%	.06
Mean number of MACE per patient per year	0.08	0.46	0.38	(–0.15, 0.90)	452%	.08
Mean number of hospitalizations per patient per year	0.08	0.27	0.19	(–0.10, 0.48)	224%	.10

Abbreviations: LDL-C, low-density lipoprotein-cholesterol; MACE, major adverse cardiac event.
[a] For patients with less a year of observation period before or after lomitapide initiation, study outcomes were annualized to estimate the annual average.
[b] The mean was calculated for patients (n=13) with LDL-C readings within 1 year before and 1 year after lomitapide discontinuation.

Figure 4: *Clinical Outcomes in Patients who Discontinued Lomitapide Treatment Following LDL-C Response (n=27)*

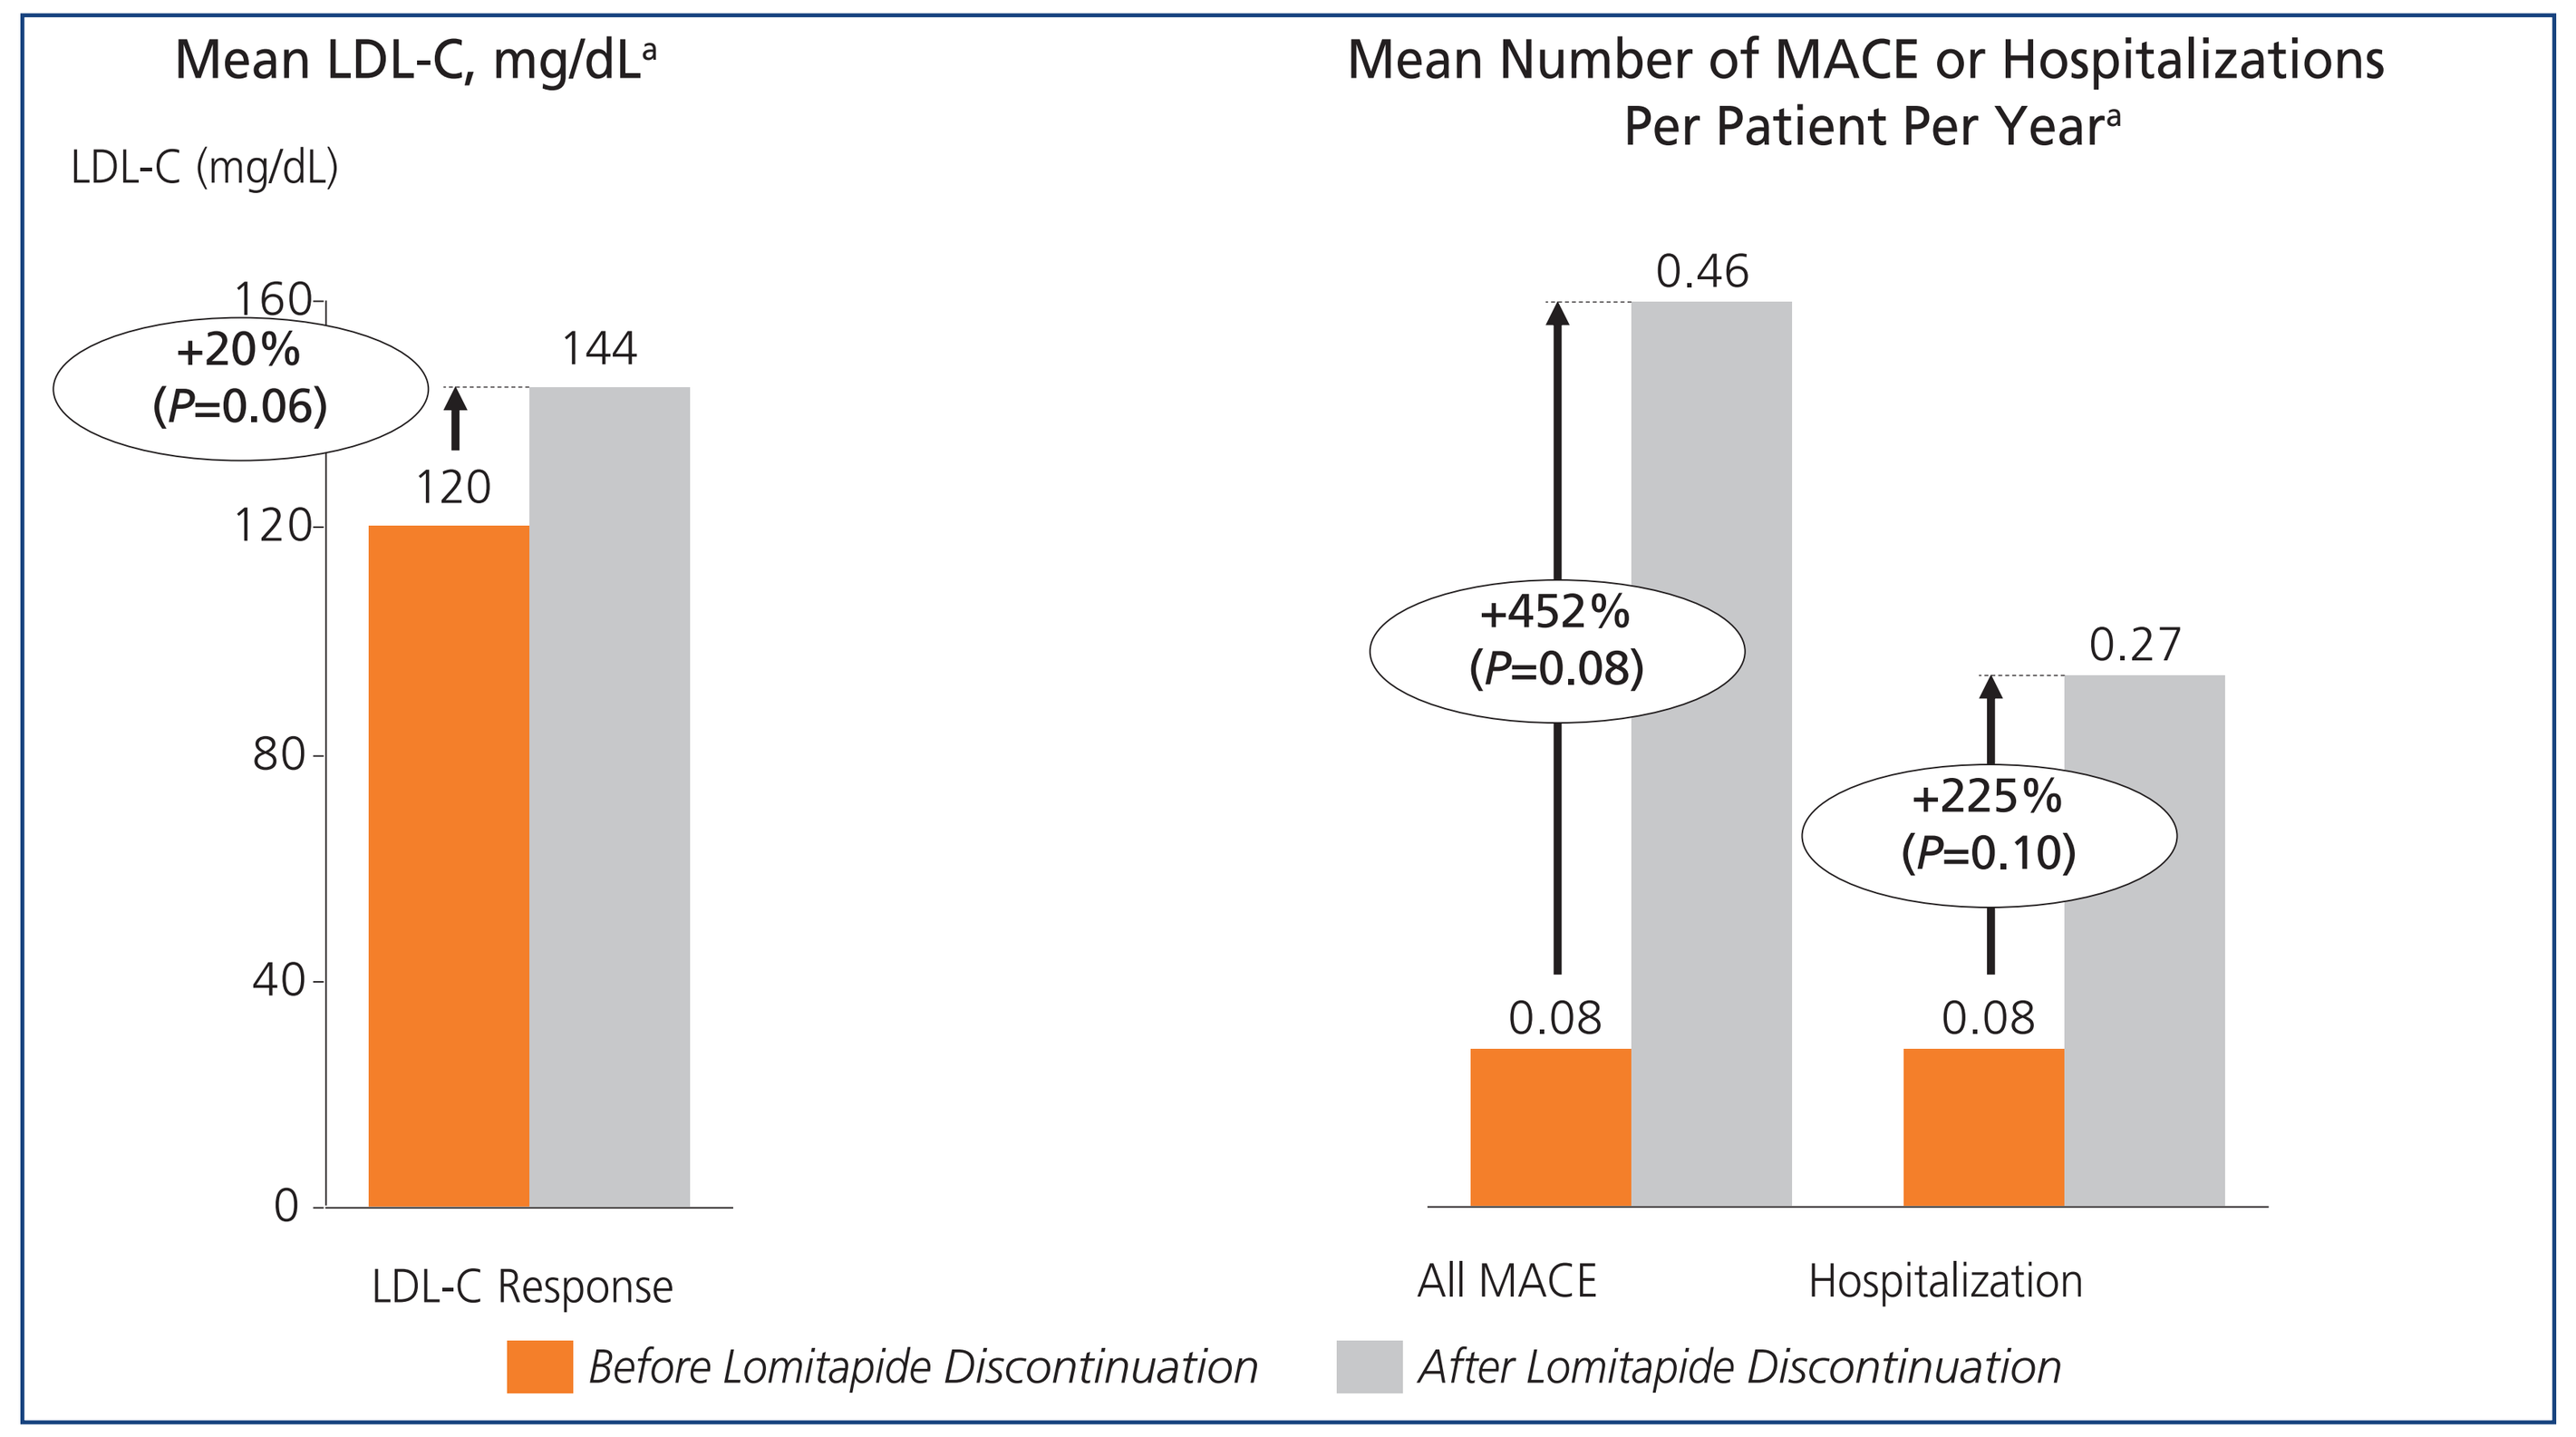
Mean LDL-C, mg/dL[a]
LDL-C (mg/dL)
160
120
80
40
0
+20%
(P=0.06)
120
144
LDL-C Response
Mean Number of MACE or Hospitalizations Per Patient Per Year[a]
0.46
+452%
(P=0.08)
0.08
0.27
+225%
(P=0.10)
0.08
All MACE
Hospitalization
Before Lomitapide Discontinuation
After Lomitapide Discontinuation

[a] All results compared in year before and after lomitapide initiation

Study Limitations

- The data are observational and therefore subject to unmeasured confounding factors. For this reason, a possibly causal impact of starting or stopping lomitapide is not established.
- Despite our use of large databases, the number of HoFH patients and the number of hospitalizations and MACE were small
- Due to the small numbers of events (MACE and hospitalizations), most of our findings (although large enough to be clinically relevant) were not statistically significant at *P*<0.05
- Our MACE and hospitalization data are from insurance claims rather than from direct reports of events, reducing confidence in our results
- Pooling of data was dependent on records from the same patient being present across multiple sources for real-world data and the need for access to these data sources

Conclusions

- This study tends to confirm that lomitapide significantly reduces LDL-C in HoFH patients in an outpatient clinical setting, consistent with clinical trials and formal registry data[3,4]
- Most importantly, this study provides evidence that there is a clinically meaningful (albeit not statistically significant) decrease in MACE and hospitalization with lomitapide treatment. These data also tend to confirm results of a recent modelling study which similarly suggested that lomitapide may reduce such events in long-term use[5]
- This study further suggests a clinically meaningful loss of decreases in LDL-C levels, MACE, and hospitalizations after lomitapide discontinuation, further supporting clinical benefit with lomitapide, and supporting a need for long-term continuation of lomitapide therapy
- The research algorithm of matching 2 separate databases at patient level is a promising approach for gathering meaningful real-world rare disease outcomes data, which may be useful in future studies of other rare diseases

Potential Conflicts of Interest

RDT, QP, and CF: Employee of Analysis Group, which received consulting fees from Aegerion Pharmaceuticals, Inc.

TS: Employee and stockholder of Aegerion Pharmaceuticals, Inc.

EAB: Consultant and/or speaker for Aegerion Pharmaceuticals, Inc., Kastle, Amgen, Regeneron, and Sanofi-Aventis

Funding Declaration

This research was funded by Aegerion Pharmaceuticals, Inc. Aegerion is the manufacturer of lomitapide.

Acknowledgments

Aegerion provided funding for medical writing support in the development of this poster; Tim Peoples and Kai Lou, employees of Analysis Group, provided editorial support for this poster. Aegerion was given the opportunity to review and provide feedback on the poster. The authors had full editorial control of the poster and provided their final approval of all content.

References

1. Cuchel M, Bruckert E, Ginsberg HN, et al. Homozygous familial hypercholesterolaemia: new insights and guidance for clinicians to improve detection and clinical management. A position paper from the Consensus Panel on Familial Hypercholesterolaemia of the European Atherosclerosis Society. *Eur Heart J.* 2014;35(32):2146–2157.
2. Cuchel M, Meagher EA, du Toit Theron H, et al. Efficacy and safety of a microsomal triglyceride transfer protein inhibitor in patients with homozygous familial hypercholesterolaemia: a single-arm, open-label, phase 3 study. *Lancet.* 2013;381(9860):40–46.
3. Blom D, Kastelein JJ, Larrey D, et al. Abstract 10818: Lomitapide Observational Worldwide Evaluation Registry (LOWER): one-year data. *Circulation.* 2015;132 (Suppl 3): A10818–A10818.
4. Blom DJ, Averna MR, Meagher EA, et al. Long-term efficacy and safety of the microsomal triglyceride transfer protein inhibitor lomitapide in patients with homozygous familial hypercholesterolemia. *Circulation.* 2017;136(3):332–335.
5. Leipold R, Raal F, Ishak J, Hovingh K, Phillips H. The effect of lomitapide on cardiovascular outcome measures in homozygous familial hypercholesterolemia: A modelling analysis. *Eur J Prev Cardiol.* 2017;24(17):1843–1850.

Presented at the 22nd World Cardiology Conference; December 11–12, 2017; Rome, Italy