Title: Efficacy and safety of oral desloratadine (10mg once daily) in patients with chronic spontaneous urticaria.

Authors: Kapadia S[1], Nageswaramma S[2], Shah K[3], Singh A[4], Mahajan SC[5], Deshpande A[6], Chakraborty S[7], Kar BR[8], Padmaja P[9], Bharija SC[10], Doshi M[11], Dhawan S[11],Gabhane M[12], Thavkar A[12], Rohira P[12], Lakhwani L[11]

[[1]Kanoria Hospital & Research Centre, Gandhinagar, India [2]Government General Hospital, Guntur, India [3]Medilink Hospital Research Centre, Ahmedabad, India [4]SMS Hospital, Jaipur, India [5]Supe Heart & Diabetes Hospital & Research Centre, Nashik, India [6]Oyster & Pearl Hospital, Pune, India [7]R.G. Kar Medical College & Hospital, Kolkata, India [8]IMS and SUM Hospital, Bhubaneswar, India [9]Osmania General Hospital, Hyderabad, India [10]Sir Ganga Ram Hospital, New Delhi, India [11]India Clinical Research, Sun Pharma, Mumbai, India, [12]India Medical Affairs, Sun Pharma, Mumbai, India]

Purpose: Chronic spontaneous/idiopathic urticaria (CSU/CIU) impairs quality of life and affects performance at work/school. Second-generation H1-antihistamines are recommended as first-line treatment for CSU. Updosing (upto 4 times) is recommended in patients unresponsive to the standard dose. This study evaluated efficacy and safety of desloratadine 10mg in the treatment of CSU. **Methodology**: A multi-centric, open-label, single-arm, phase 3 study was conducted in India in adult patients with moderate-to-severe CSU, active CSU ≥3 weeks, wheals present for ≥3 days/week and not responding to ≥2 weeks of desloratadine 5mg . Desloratadine 10mg was administered orally once daily for 4 weeks. Study assessments included weekly Urticaria Activity Score (UAS7) and Chronic Urticaria Quality-of-Life Questionnaire (CU-Q2oL). **Results:** Total 256 patients (mean age: 35.3 years) were enrolled in the study. Desloratadine 10mg led to a significant improvement as early as at week 2. Significant improvement by 42.32% ($p<0.0001$) was seen in mean UAS7 at week 4 (18.17) compared to baseline (31.90). The mean CU-Q2oL score improved by 38.48% from baseline (59.81) to week 4 (35.36) ($p<0.0001$). By week 4, 21% of patients improved to mild urticaria (UAS7=7-15) and 10% of patients were either well controlled or urticaria free (UAS7=0-6). Adverse events reported in 5.5% of patients were mostly mild (80%), non-serious and did not lead to discontinuation. Two events (0.8%) of dry mouth were reported. Somnolence/drowsiness was not reported in this study. **Conclusion:** Desloratadine 10mg once daily was efficacious and well tolerated in patients with moderate-to-severe CSU nonresponsive to standard dose desloratadine. (Study registration number: CTRI/2017/10/010146)

Biography

Dr Mukesh Gabhane has completed his graduation in Bachelor of Medicine and Bachelor of Surgery (MBBS) from the Maharashtra University of Health Sciences (MUHS), Nashik, followed by MBA (pharmaceuticals) from Narsee Monjee Institute of Management Studies, Mumbai. He has experience of over a decade working in medical affairs (medico-marketing, pharmacovigilance and clinical studies).

Eail id: mukesh.gabhane@sunpharma.com

Presenting author details

Full name: Dr Mukesh Gabhane

Contact number: +919619101705

Twitter account: @MukeshGabhane

LinkedIn account: https://www.linkedin.com/in/mukesh-gabhane-56710833

Session name/number: Urticaria and angioedema/19

Category: E poster presentation